Poster 1765

AO-176, A Humanized Anti-CD47 Monoclonal Antibody that Directly Kills Human Tumor Cells and has Additional Unique Functional Characteristics

Robyn Puro, Katherine Liu, Benjamin Capoccia, Michael Donio, Ronald Hiebsch, Myriam Bouchlaka, Alun Carter, Pamela Manning, Kathleen Crowley and Robert Karr

Arch Oncology, 4320 Forest Park Avenue, St. Louis, MO 63108 and 2000 Sierra Point Parkway, Brisbane, CA 94005

Introduction

CD47 is a cell surface glycoprotein that interacts with signal regulatory protein alpha (SIRPα) on macrophages and dendritic cells triggering a “don’t eat me” signal that inhibits phagocytosis. Many tumors evade immune surveillance by overexpressing CD47, thereby preventing their recognition by phagocytes. Blocking the interaction of SIRPα/CD47 promotes phagocytosis and tumor cell destruction leading to a reduction in tumor burden.

We have developed a humanized anti-CD47 antibody, AO-176, that blocks the interaction between CD47 and SIRPα to stimulate tumor cell phagocytosis but also exhibits several additional novel functional characteristics. One of these characteristics is the ability of AO-176 to directly induce cell death in multiple human tumor cell lines by a cell autonomous mechanism (not ADCC). A second novel characteristic exhibited by AO-176 is its selective binding to tumor cells compared to normal cells including red blood cells (cynomolgus monkey and human), endothelial, epithelial and skeletal muscle cells. The reduced binding to red blood cells (RBCs) is expected to not only diminish their ability to act as an AO-176 sink but also to minimize on target toxicity observed in patients following treatment with other RBC-binding CD47 antibodies. A third novel characteristic is enhanced binding to tumor cells at acidic pH. AO-176 binds to human tumor cell lines in the high pM to low nM range at physiologic pH, however, binding is enhanced up to 20-fold at an acidic pH of 6.5. An acidic pH of 6.4-7.2 is characteristic of the microenvironment of solid tumors and correlates with tumor progression and metastasis. As a result of this enhanced binding at acidic pH, AO-176 has the potential added advantage of tumor-specific targeting. Combined with the ability of AO-176 to induce tumor phagocytosis, these novel characteristics have the potential to enhance AO-176 efficacy while reducing typical anti-CD47 antibody side effects. Importantly, dose-dependent efficacy has been demonstrated in multiple mouse tumor xenograft models.

Taken together, the unique combination of functional characteristics of AO-176, including induction of cell-autonomous killing, enhanced binding to tumor cells at acidic pH, significantly reduced binding to normal cells and potent *in vivo* efficacy provides the preclinical rationale for further development and differentiates this antibody from others in development.

AO-176: A Humanized anti-CD47 mAb

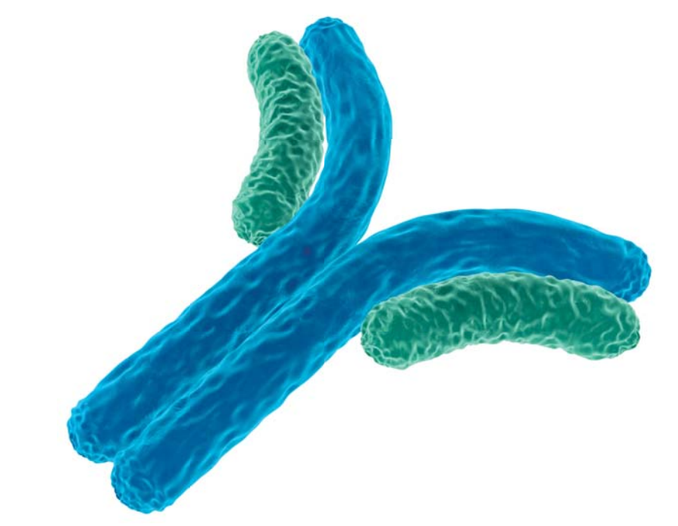

- Humanized IgG2
- Selectively binds to human CD47 on tumor cell lines (EC_{50} 1.5 – 32 nM)
- Negligible binding to human RBCs
- Reduced binding to normal cells
- Novel DAMP induction and direct killing of tumor cells (non-ADCC)
- Greater affinity at acidic pH (potential tumor targeting mechanism)
- Blocks CD47/SIRPα interaction to induce phagocytosis of tumor cells
- Anti-tumor efficacy in human xenograft models
- Minimal impact on red cells in preliminary non-GLP toxicology studies

Mechanism of Action: Blocking, DAMP Induction and Killing

CD47/SIRPα

Tumor Cell Immune Evasion “Don’t Eat Me Signal”

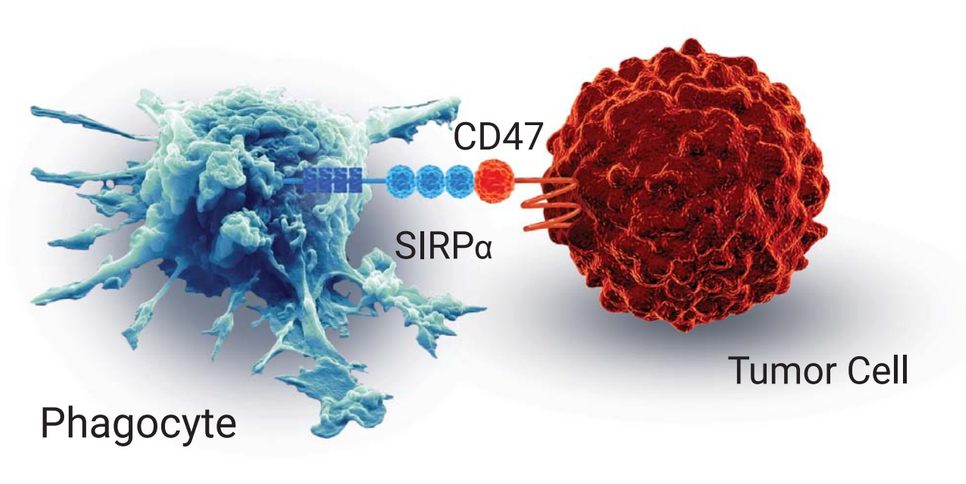
CD47
SIRPα
Phagocyte
Tumor Cell

- CD47/SIRPα binding conveys ‘don’t eat me’ signal to phagocytes
- Elevated CD47 expression enables tumors to escape innate immune surveillance

Blocking

Blocking: AO-176 Promotes Phagocytosis of Tumor Cells

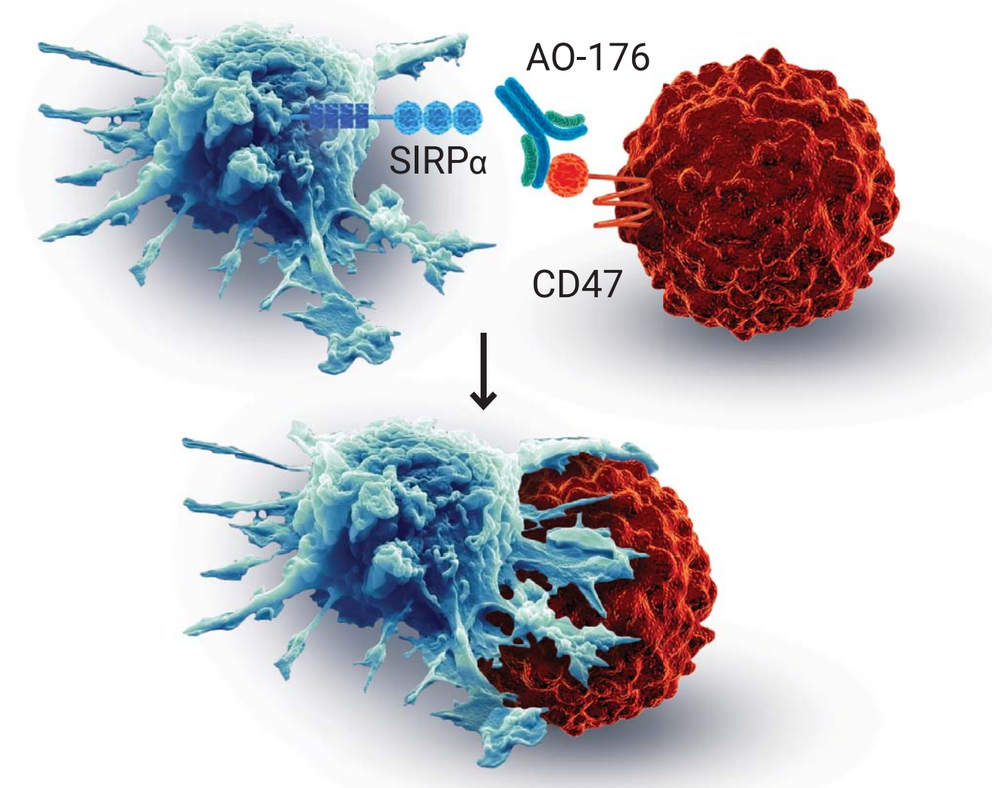
AO-176
SIRPα
CD47

- AO-176 disrupts checkpoint inhibition

DAMPs and Killing

AO-176 Stimulates DAMPs and Directly Targets AND Kills Tumor Cells

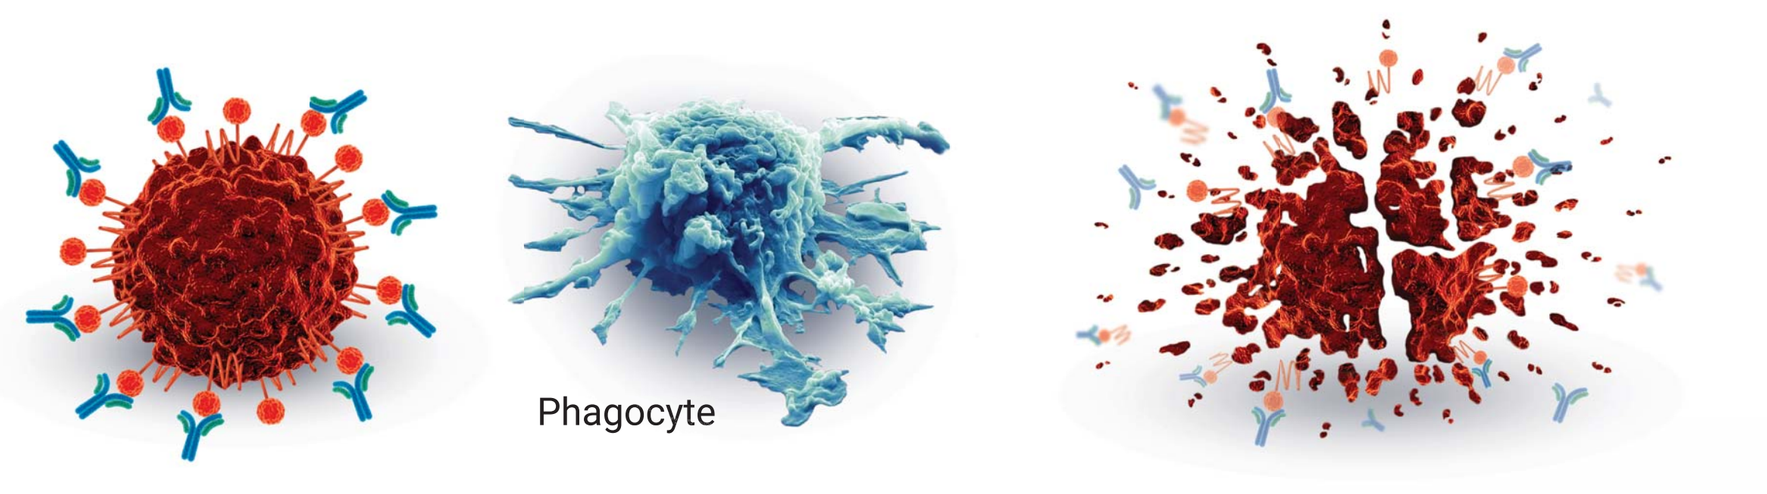
Phagocyte

- AO-176 binds to tumor cell
- AO-176 induces cellular stress (AV+/7AAD-) and causes the release and exposure of Damage Associated Molecular Patterns (DAMPs) by tumor cells (immunogenic-like cell death)
- AO-176 directly kills tumor cells (AV+/7AAD+) but not normal cells

AO-176 Preferentially Binds Tumor vs. Normal Cells

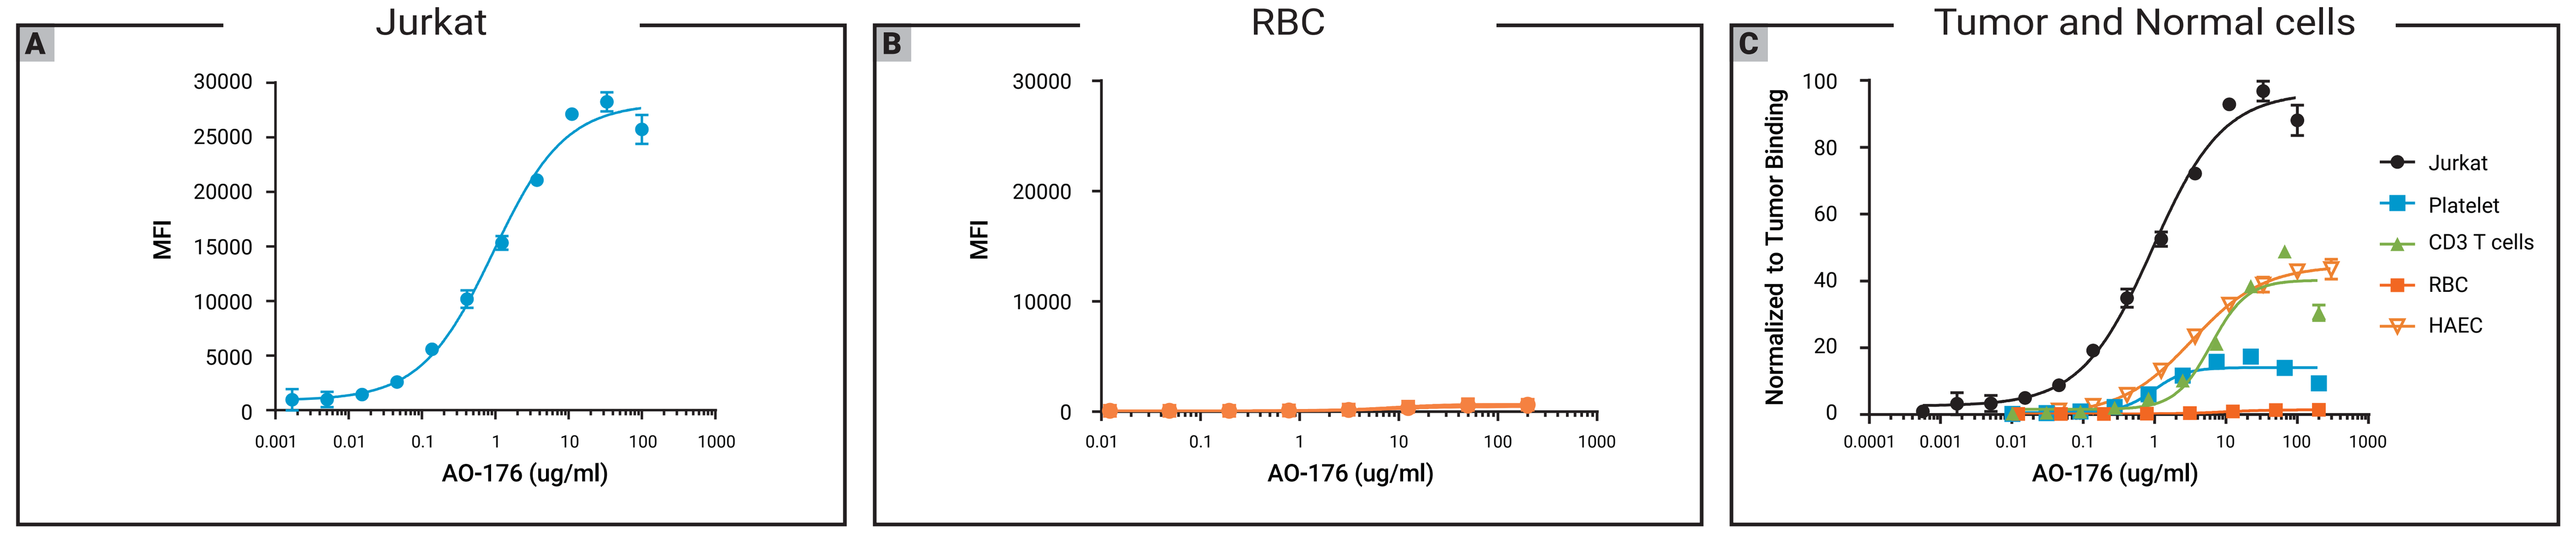
A
Jurkat
MFI
AO-176 (ug/ml)
B
RBC
MFI
AO-176 (ug/ml)
C
Tumor and Normal cells
Normalized to Tumor Binding
AO-176 (ug/ml)
Jurkat
Platelet
CD3 T cells
RBC
HAEC

Binding of AO-176 to human CD47 on (A) Jurkat cells or (B) to freshly isolated human red blood cells (RBCs) determined using flow cytometry measuring mean fluorescence intensity (MFI). Increasing concentrations of AO-176 were added to cells followed by incubation with a fluorescently-labeled secondary antibody. (C) Comparison of binding of AO-176 to tumor cells with reduced binding to human platelets, CD3 T cells, RBC and human aortic endothelial cells.

AO-176 Blocks the Binding of CD47 to SIRPα and Promotes Tumor Cell Phagocytosis

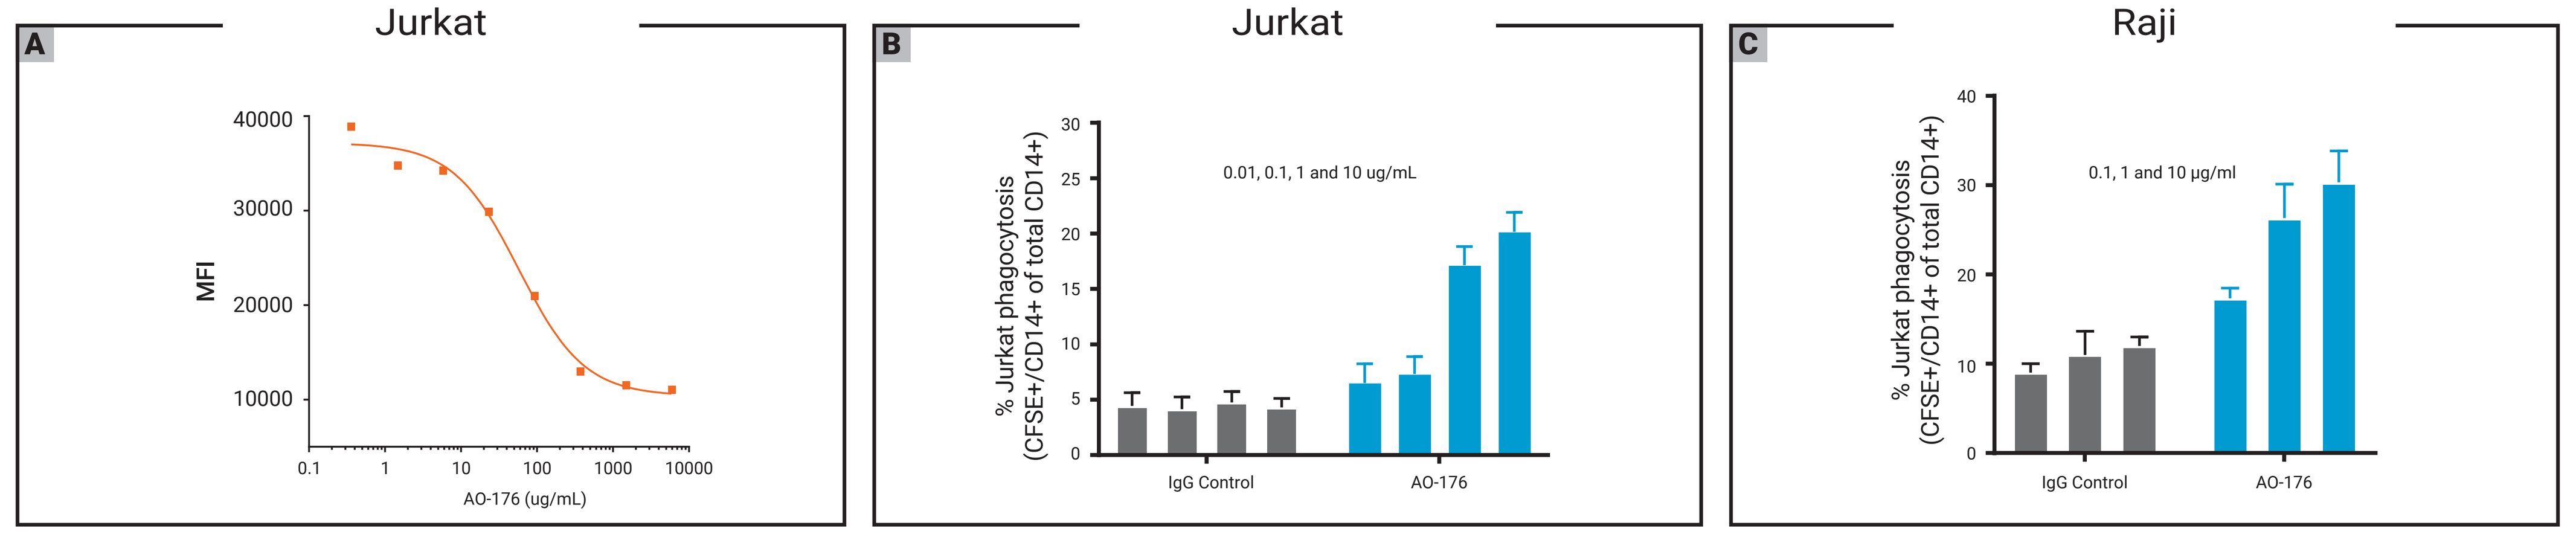
A
Jurkat
MFI
AO-176 (ug/mL)
B
Jurkat
% Jurkat phagocytosis (CFSE+/CD14+ of total CD14+)
0.01, 0.1, 1 and 10 ug/mL
IgG Control
AO-176
C
Raji
% Jurkat phagocytosis (CFSE+/CD14+ of total CD14+)
0.1, 1 and 10 µg/ml
IgG Control
AO-176

(A) Inhibition of CD47 binding to SIRPα by AO-176 was assessed in vitro using a fluorescently-labeled SIRPα-Fc fusion protein. Jurkat cells were incubated with AO-176 (0.3 – 20 µg/mL) and fluorescently-labelled SIRPα-Fc fusion protein and inhibition was determined by measurement of labelled SIRPα-Fc signal (mean fluorescence intensity shown). For phagocytosis assays, PBMC derived macrophages were co-cultured with CFSE-labeled tumor cells ± AO-176 or Control IgG. AO-176 promotes phagocytosis of both (B) Jurkat and (C) Raji cells.

AO-176 Mediates Cell-autonomous Early and Late Apoptotic Killing of Tumor Cells

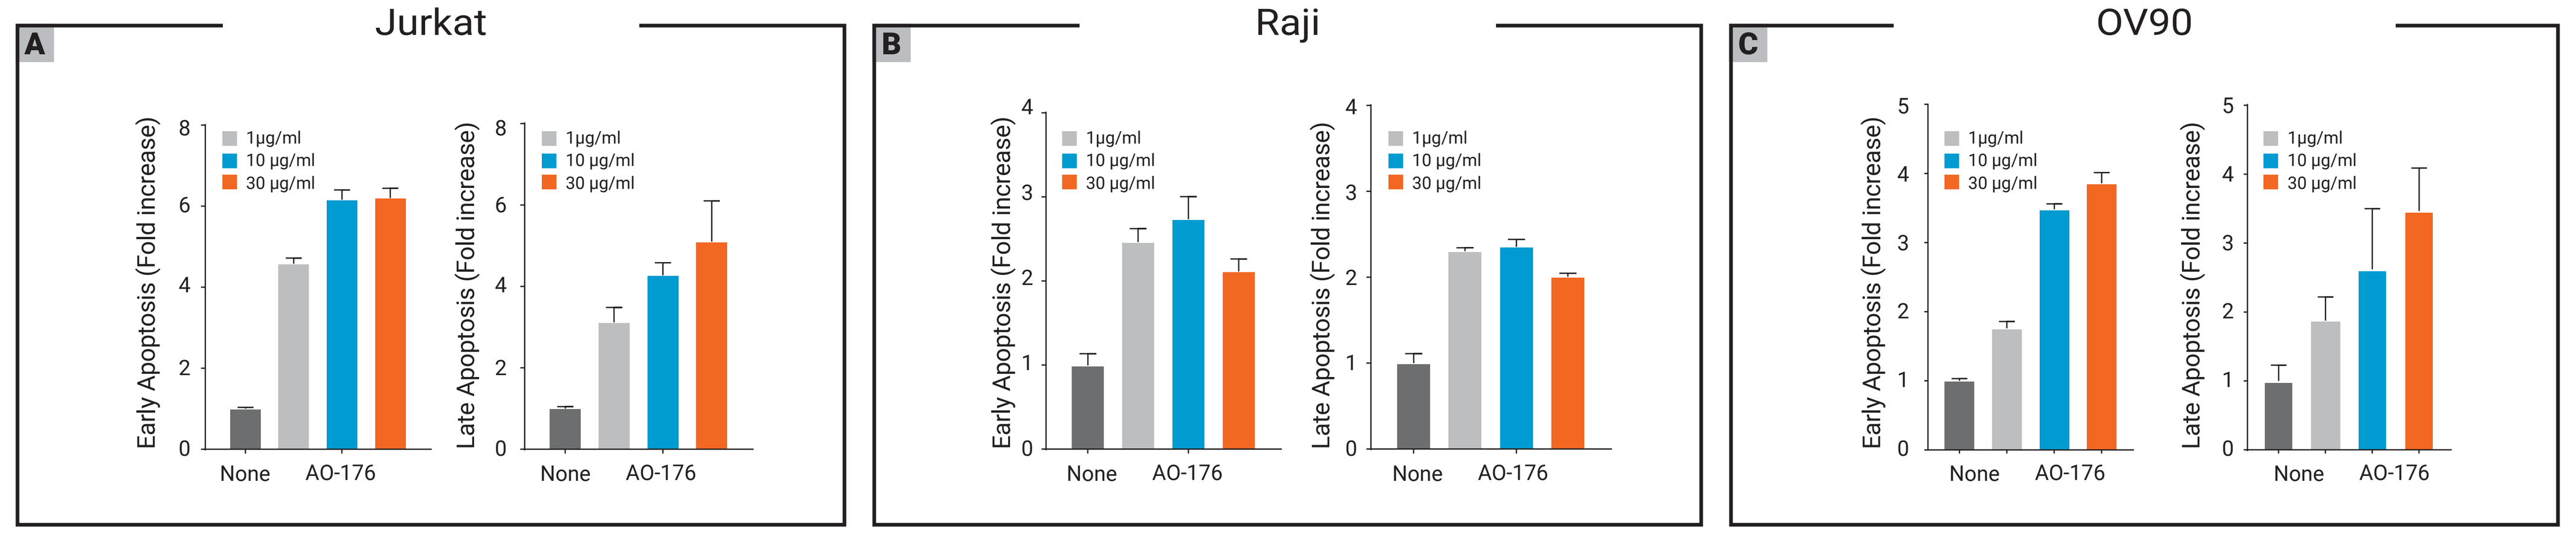
A
Jurkat
B
Raji
C
OV90
Early Apoptosis (Fold increase)
Late Apoptosis (Fold increase)
1µg/ml
10 µg/ml
30 µg/ml
None
AO-176

Cell autonomous killing by AO-176 was measured in (A) Jurkat (B) Raji B lymphoma or (C) OV90 ovarian carcinoma cells incubated with various concentrations of AO-176 for 24 hours at 37°C. Cells were stained with Annexin V to measure externalization of phosphatidyl serine (Annexin V+), a marker of early apoptosis, and uptake of 7-AAD dye, an indicator of apoptotic cell death. Signal intensity of Annexin V+ (AV+) and 7AAD+ cells was detected by flow cytometry. The data are shown as fold-change in early apoptotic (Annexin V+/7AAD negative) over untreated cells (none) and fold-change late apoptotic (Annexin V/7-AAD double positive) over untreated cells.

AO-176 Induces DAMP Signaling

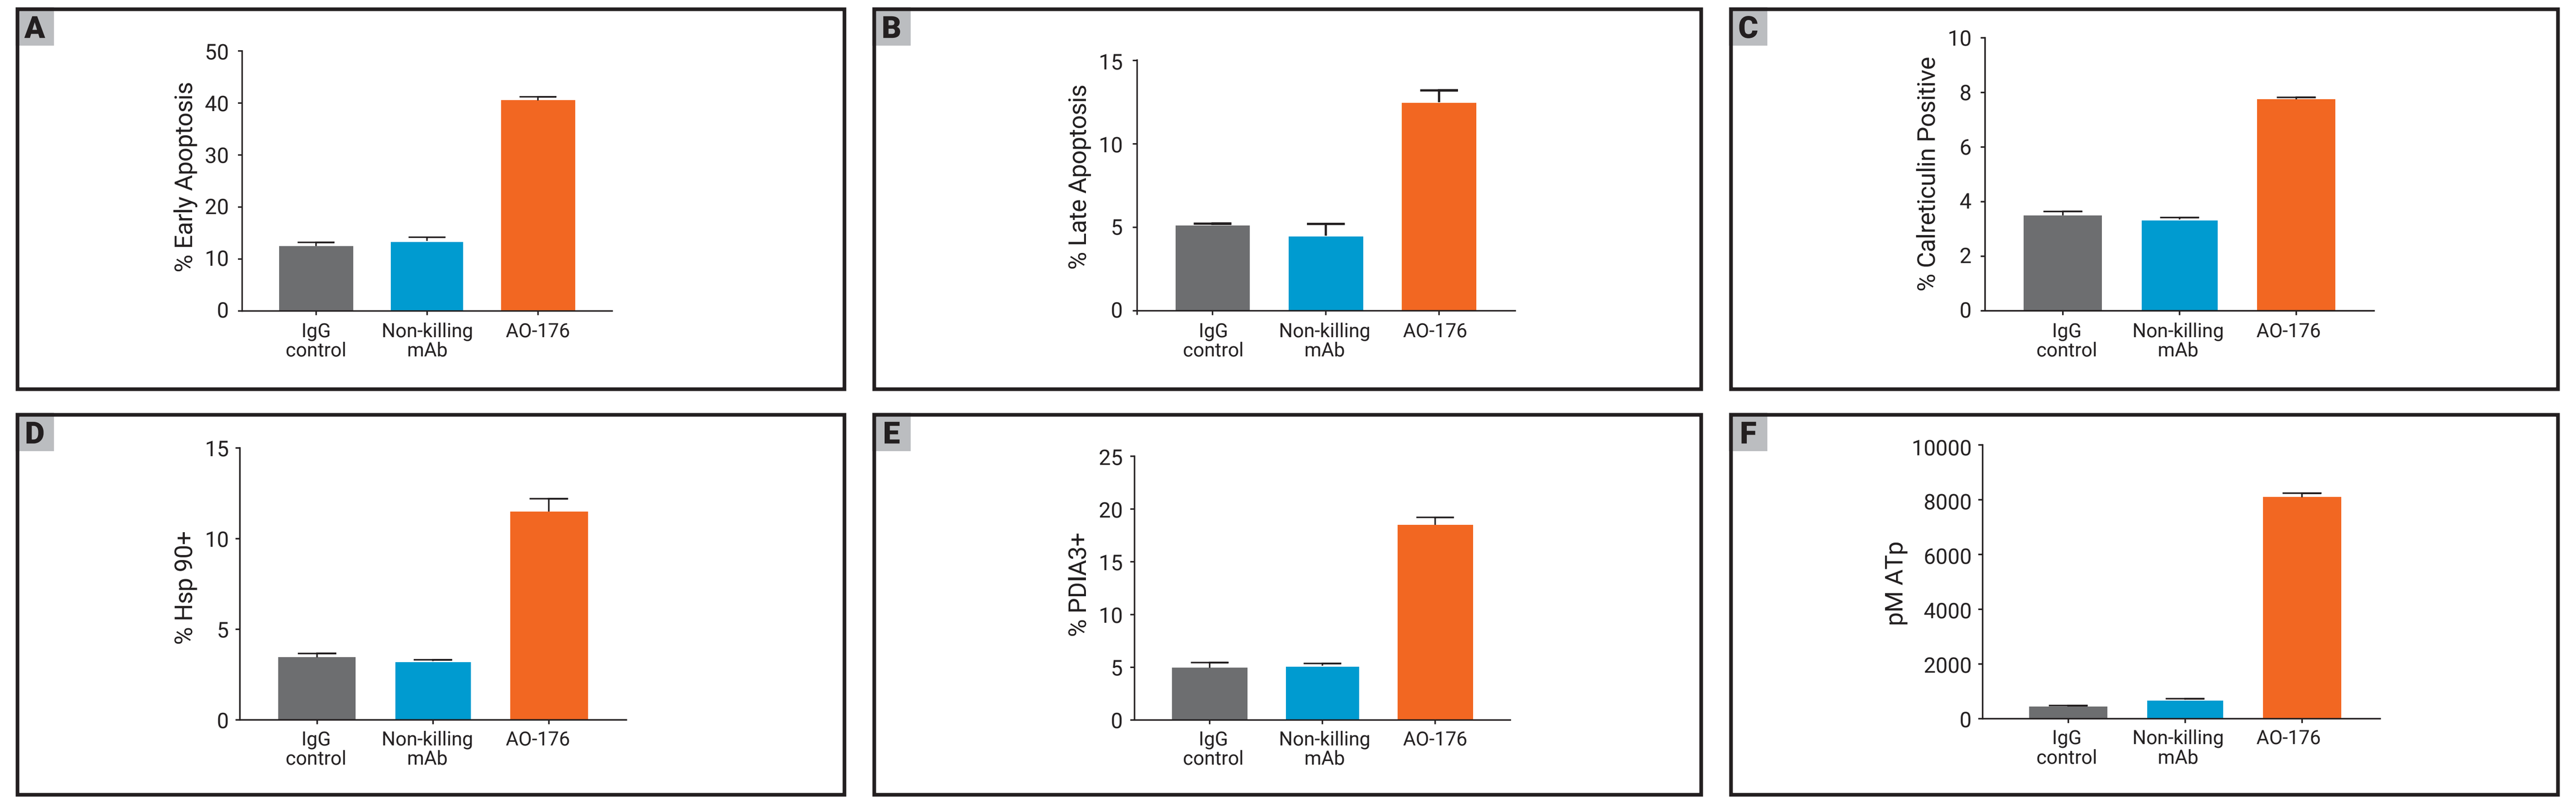
A
% Early Apoptosis
B
% Late Apoptosis
C
% Calreticulin Positive
D
% Hsp 90+
E
% PDIA3+
F
pM ATp
IgG control
Non-killing mAb
AO-176

Induction of Danger Associated Molecular Patterns (DAMPs), endogenous danger signals which are markers of cell stress and promote inflammatory responses, were assessed in Jurkat cells. Addition of AO-176 but not a non-killing anti-CD47 mAb that both binds with high affinity to CD47 and causes phagocytosis, induced DAMP signaling as detected by externalization of (A) phosphatidyl serine (AV+), (C) calreticulin (CRT+), (D) heat shock protein 90 (HSP90+) and (E) Protein Disulfide Isomerase Family A Member 3 (PDIA3), measured by flow cytometry and release of (F) ATP measured by ELISA. AO-176 also induced cell death (B) as measured by both externalization of phosphatidyl serine (early apoptosis) and uptake of 7-AAD dye (AV+ 7AAD+, late apoptosis).

AO-176 Binding to CD47 Expressed on Tumor Cells is Enhanced at Acidic pH

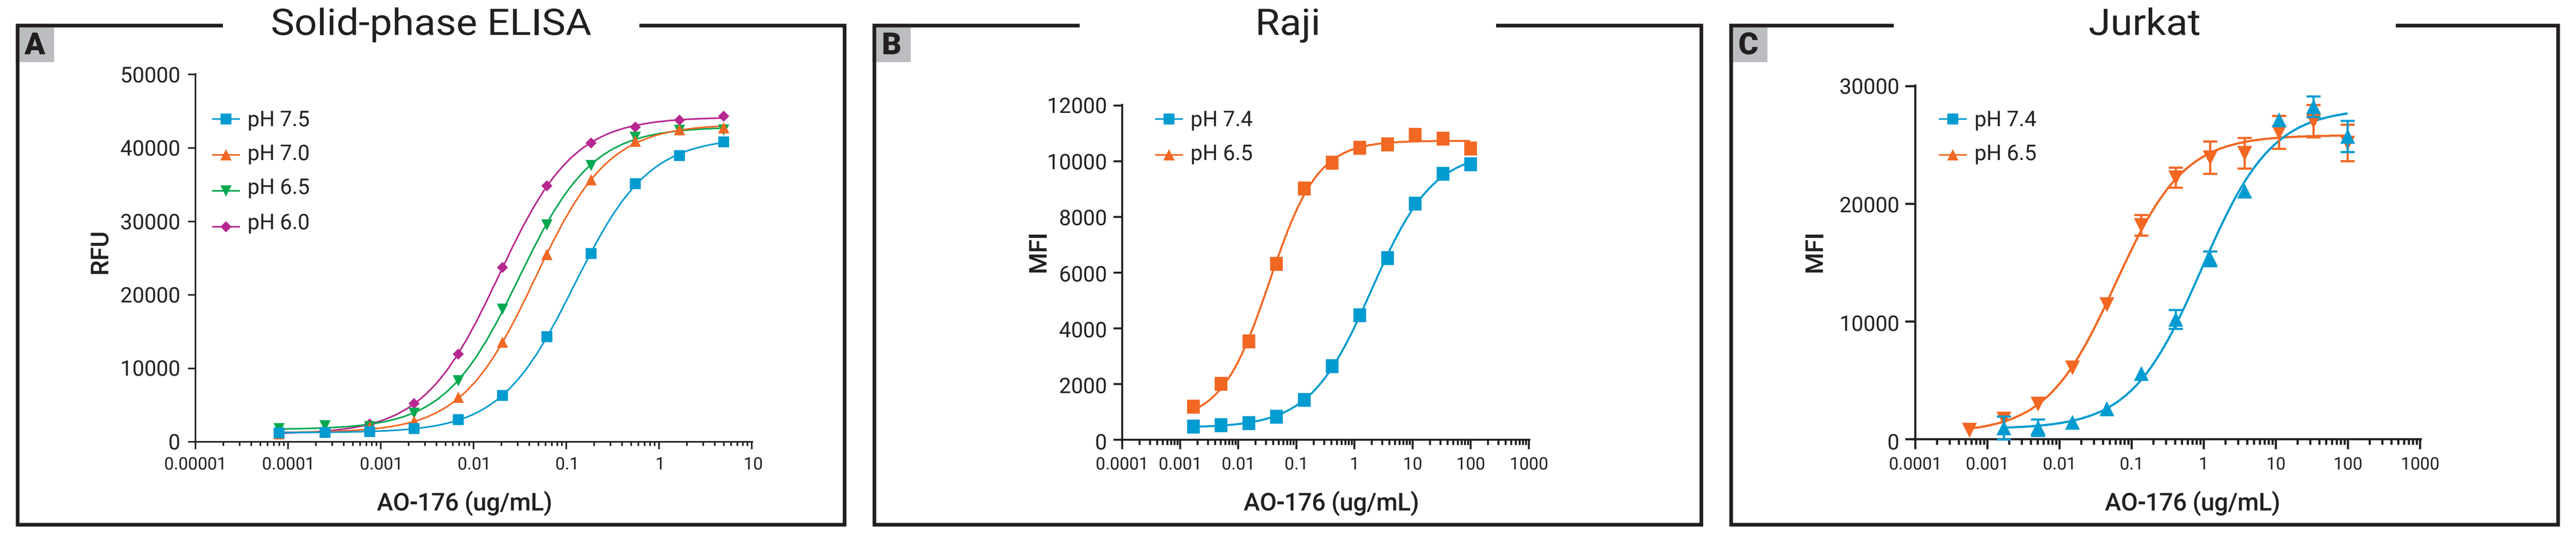
A
Solid-phase ELISA
RFU
pH 7.5
pH 7.0
pH 6.5
pH 6.0
AO-176 (ug/mL)
B
Raji
MFI
pH 7.4
pH 6.5
AO-176 (ug/mL)
C
Jurkat
MFI
pH 7.4
pH 6.5
AO-176 (ug/mL)

pH dependent binding of AO-176 to human CD47 was measured by solid-phase ELISA (A; relative fluorescence units) and flow cytometry (B, C; mean fluorescence intensity) on Raji B-Cell lymphoma cells and Jurkat Cells. Increasing concentrations of AO-176 were added to plates coated with recombinant human CD47 extracellular domain or to tumor cells followed by incubation with either HRP-labeled or fluorescent-labelled secondary antibody.

AO-176 is Efficacious in the Raji Tumor Xenograft Model

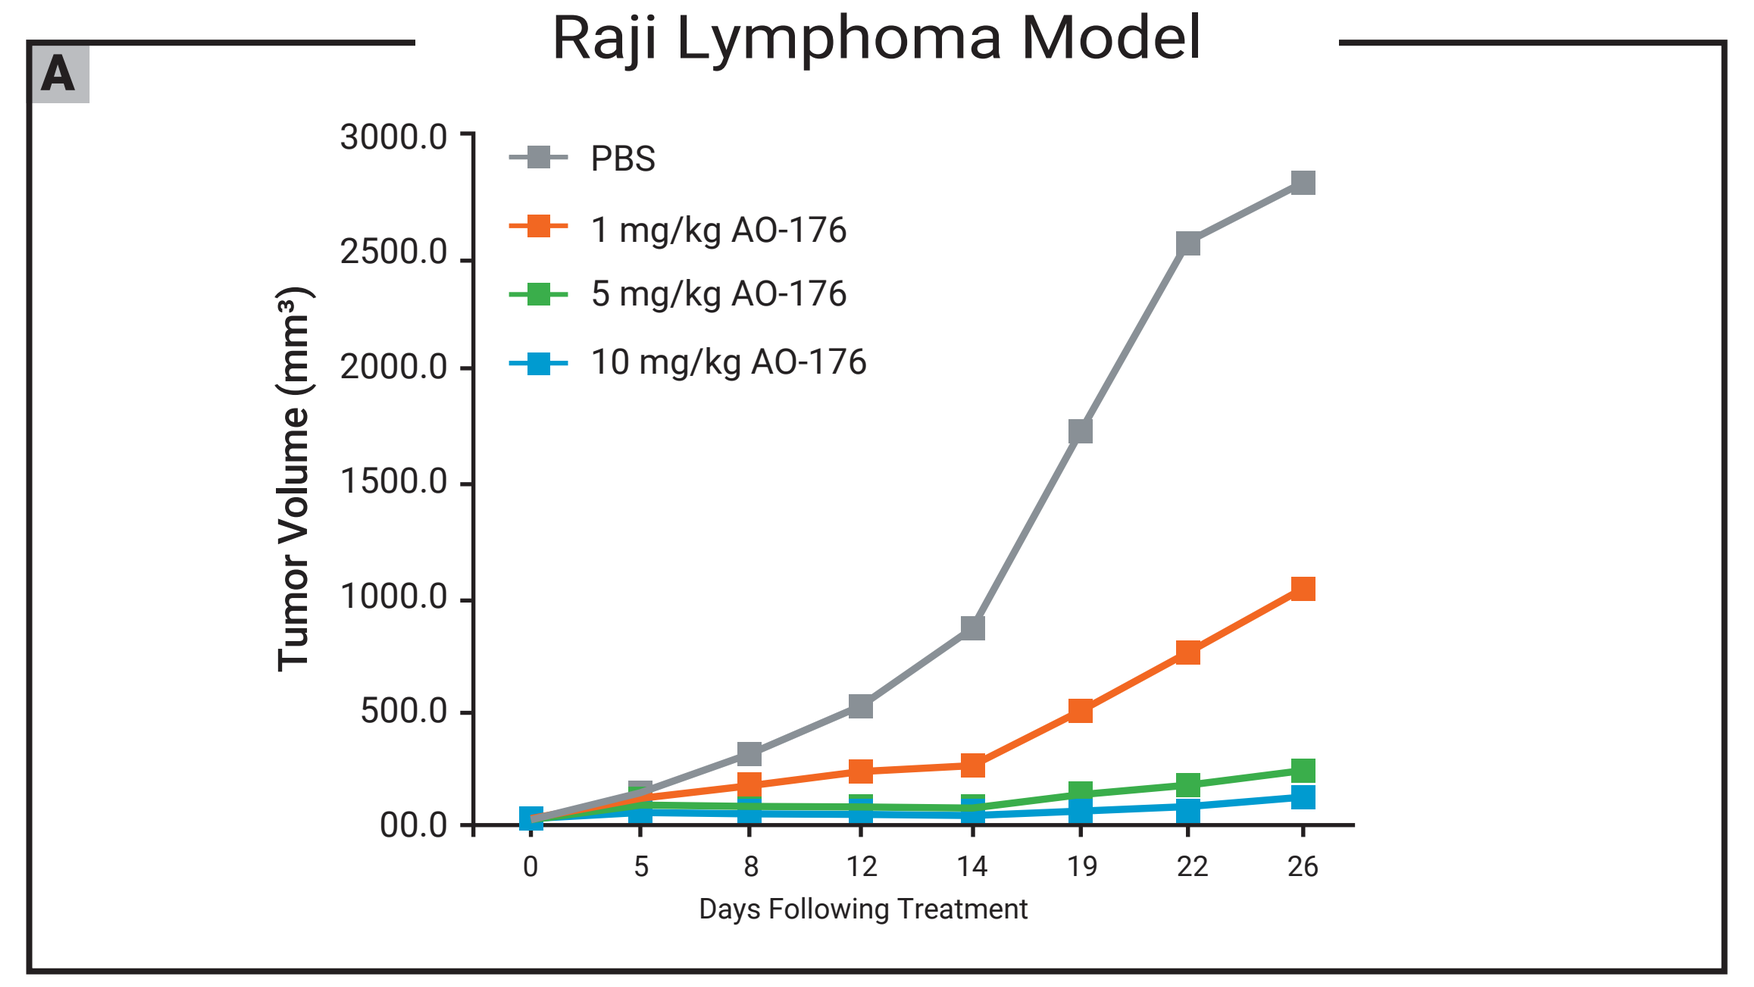
A
Raji Lymphoma Model
Tumor Volume (mm³)
PBS
1 mg/kg AO-176
5 mg/kg AO-176
10 mg/kg AO-176
Days Following Treatment

AO-176 Minimally Impacts RBC and Hemoglobin in Cynomolgus Monkey

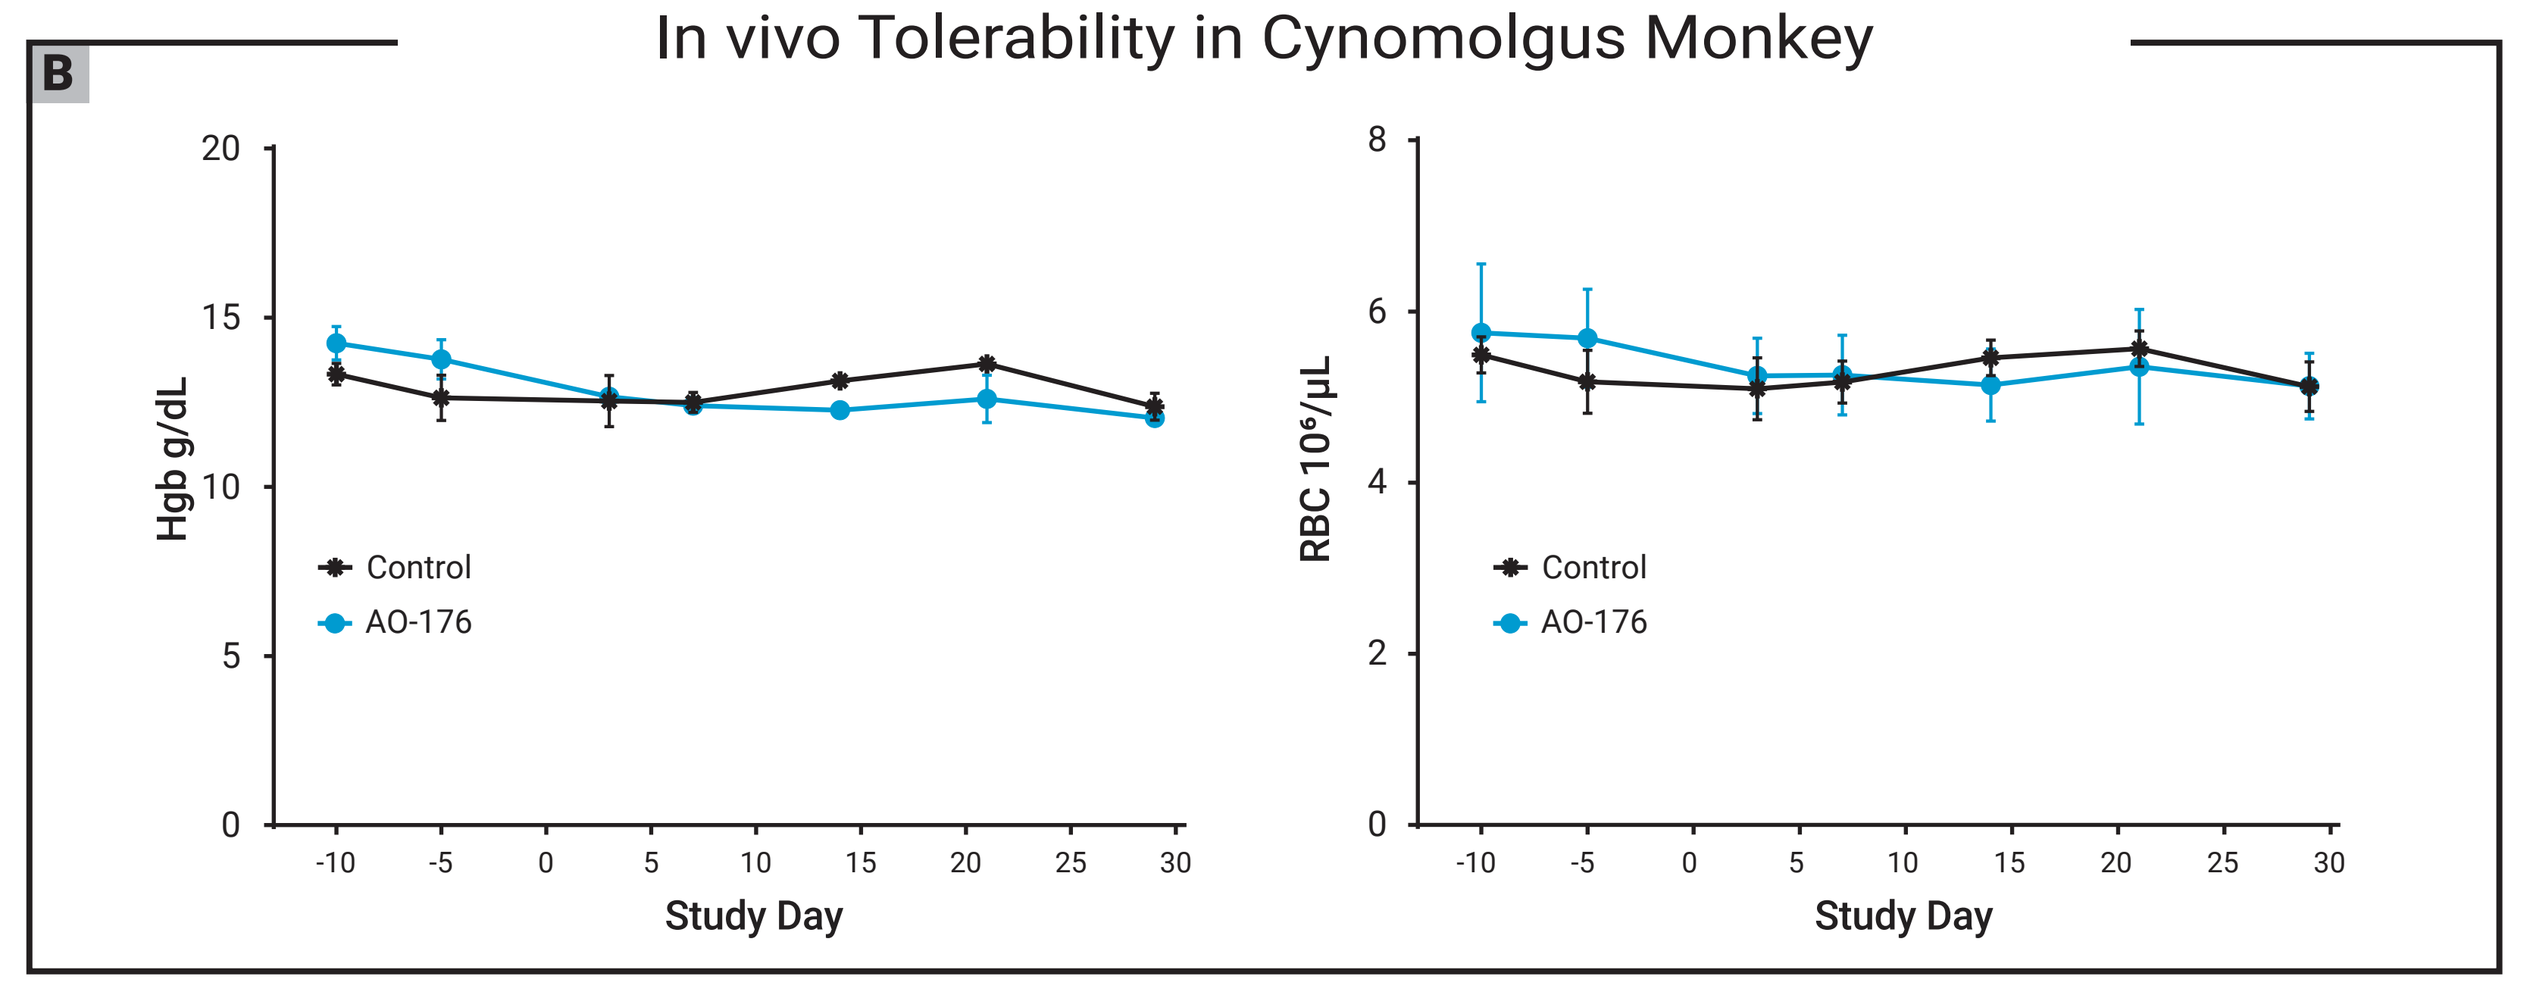
B
In vivo Tolerability in Cynomolgus Monkey
Hgb g/dL
RBC 10⁶/µL
Control
AO-176
Study Day

Human Raji B cell lymphoma cells were injected subcutaneously. When tumors reached ~50 mm^3, AO-106 (at doses from 1 mg/kg to 10 mg/kg) was administered 5X/week by intraperitoneal injection for 5 weeks. Tumor volume was measured twice/week. Treatment with AO-106 showed potent, dose-dependent tumor reduction (A). A0-176 tolerability was examined in a four-week exploratory safety study in cynomolgus monkeys, where drug was given as weekly IV infusions (first dose of 5 mg/kg followed by 3 weekly doses of 50 mg/kg). AO-176 treatment resulted in minimal reduction in red cells and hemoglobin (B). These data are consistent with the reduced RBC binding observed with AO-176.

Conclusions

AO-176 is an anti-CD47 antibody with unique differentiating characteristics

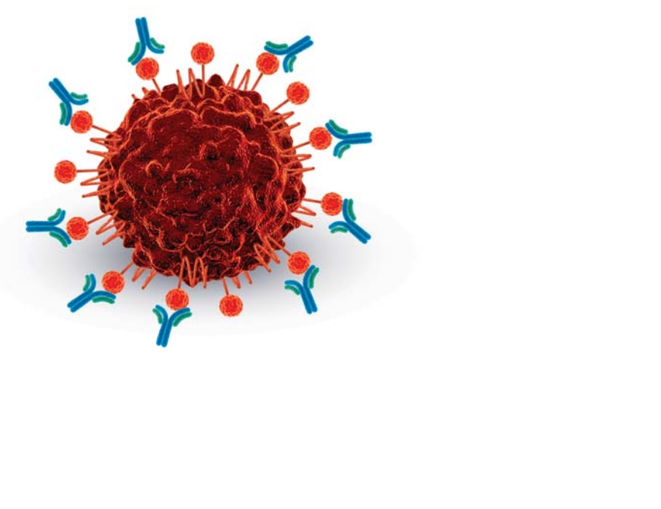

1. **Mechanism of Action:** Blocking and Direct Killing
2. **Preferential Binding** to Tumor Cells and Not Normal Cells
3. **Potential for Better Binding** in Tumor Environment (low pH)

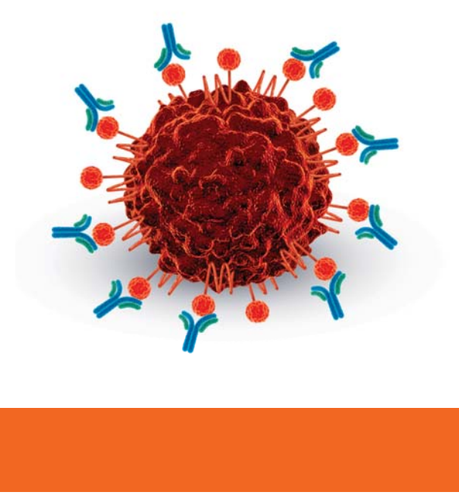

Acknowledgements

- This study was supported by Arch Oncology. The authors thank D. Cook for editorial assistance.